Digital Biology for Better Antibody-Drug Conjugates – Modelling Cell-Level Pharmacokinetics

Tatu Lindroos[1], Stanley Sweeney-Lasch[2], Stefan Hecht[2], Michael Krug[1]

Computational Chemistry & Biology[1], ADC Design and Characterization [2]
Discovery Technologies, Merck KGaA, Frankfurter Str. 250, 64293 Darmstadt, Germany

Antibody drug conjugates (ADCs) are therapeutic molecules that are aimed to deliver highly potent cytotoxic agents specifically to tumor cells with the help of a tumor-targeting monoclonal antibody that is linked to the payload drug. In a cell-level view, the ADCs bind to antigens expressed on the surface of the tumor cell. The ADC-antigen complexes get internalized, the linkers are cleaved, and the drug released. However, the ADC can already deconjugate outside the cell, depending on the properties of the linker and the drug. Free payload drug can also influx and efflux into and out of the cell. All these processes can be described mathematically with rate constants and differential equations. Accurate quantitative characterization of cellular ADC disposition could help in understanding and improving design of ADCs.

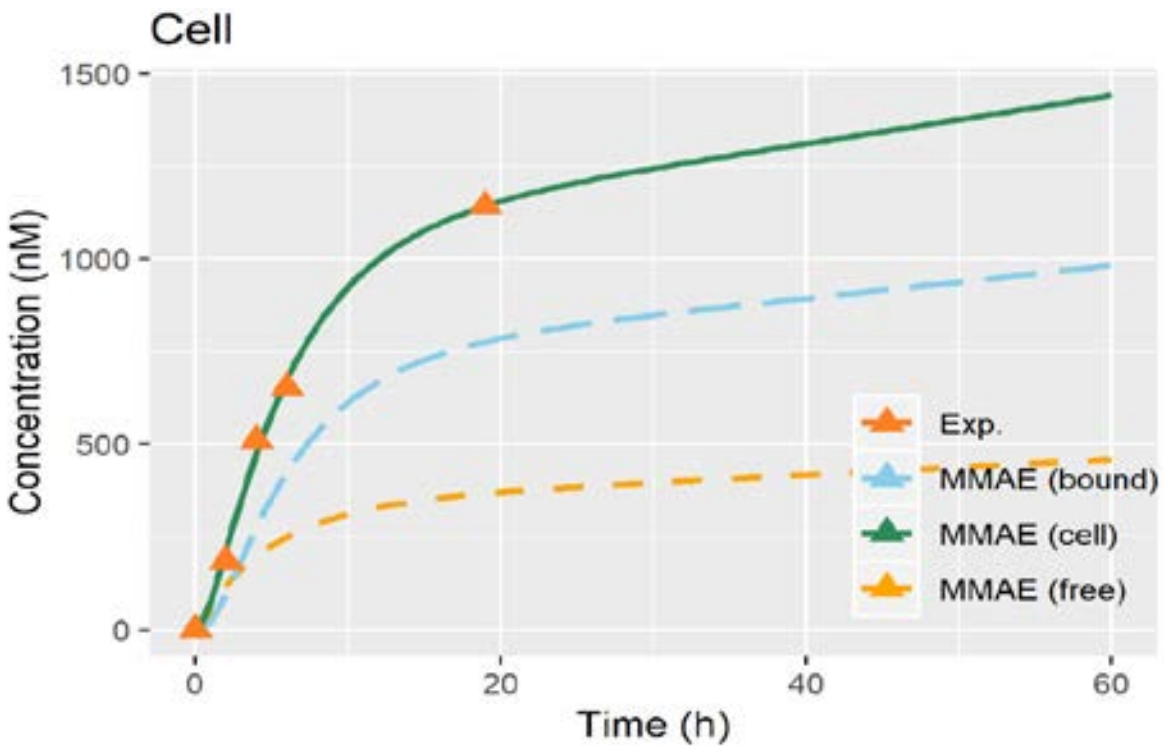
Cell
1500
1000
500
0
Concentration (nM)
0
20
40
60
Time (h)
Exp.
MMAE (bound)
MMAE (cell)
MMAE (free)

We used a model by Singh and Shah [1], built for trastuzumab-valine-citrulline-monomethyl auristatin E (Trastuzumab-vc-MMAE), to explore how different ADC constructs might behave in a cell-level system. Taking internal experimental data from an uptake assay of MMAE and from measuring the internalization of trastuzumab-vc-MMAE, we could fit the model to the ADCs used at Merck. Extending the model to also handle cell killing lead us to link ADC properties, like drug-antibody ratio and permeability, to predicted IC50 values.

The model was implemented with R Shiny library which gave it an easy-to-access browser interface and deployed on Docker container platform which enabled experimental scientists to use the tool from anywhere in the intranet. The project source code was managed in company’s internal GitLab to aid collaborative development, and a one-button build-and-deploy pipeline enabled rapid prototyping with fast feedback from the users.

[1] A. P. Singh, D. K. Shah, *Drug Metab. Dispos.*, **2017**, *45*, 1120-1132.